NSABP B-59/GBG 96-GeparDouze: A randomized double-blind phase III clinical trial of neoadjuvant chemotherapy (NAC) with atezolizumab or placebo in patients (pts) with triple-negative breast cancer (TNBC) followed by adjuvant atezolizumab or placebo

CE Geyer, Jr,[1,2] S Loibl,[3] P Rastogi,[1,4,5] S Seiler,[3] JP Costantino,[1,5] V Nekljudova,[3] P Cortazar,[6] PC Lucas,[1,5] C Denkert,[3,7] EP Mamounas,[1,8] C Jackisch,[3,9] N Wolmark[1,10]

[1]NSABP Foundation; [2]Virginia Commonwealth University Massey Cancer Center; [3]German Breast Group (GBG Forschungs GmbH); [4]Magee-Womens Hospital of UPMC; [5]University of Pittsburgh; [6]Genentech, Inc.; [7]Institut für Pathologie, Philipps-Universität Marburg und Universitätsklinikum Marburg; [8]Orlando Health UF Cancer Center; [9]Sana Klinikum Offenbach GmbH; [10]Allegheny Health Network Cancer Institute

Characteristics of TNBC Supporting Evaluation of Immune Therapies in Early Breast Cancer

- Heterogeneous group of cancers characterized by
 - High-grade tumors
 - <1% of cells + for ER and PR receptors
 - Negative for HER2 amplification or overexpression
- TNBC is associated with higher percentages of pCR to neoadjuvant chemotherapy (NAC), and women with a pCR have a favorable prognosis
- Patients (pts) with residual TNBC following NAC have higher risk for recurrence than pts with other subtypes of BC with residual disease (1,2)
- Poor survival once metastatic disease develops
- Increased incidence and worse outcomes in women of African descent

1) Liedtke C. J Clin Oncol 2008; 26: 1275-81
2) Hahnen E. JAMA Oncol. 2017; 3: 1378-85

Background

- Therapeutic blockade of PD-L1 binding by atezolizumab has resulted in relevant anti-tumor efficacy in TNBC (1,2,3)
- The Phase III IMpassion130 study in first-line metastatic TNBC reported the addition of atezolizumab to nab-paclitaxel prolonged PFS from 5.5 to 7.2 mos (HR=0.80; p=0.0025) in the overall population and from 5.0 to 7.5 mos (HR=0.62; p<0.001) in the PD-L1 positive cohort (HR=0.62). Median OS was improved from 17.6 to 21.3 mos (HR=0.84; p=0.08) overall and was improved from 15.5 to 25.0 mos in the PD-L1 positive cohort (HR=0.62) (4)
- Addition of pembrolizumab to neoadjuvant paclitaxel increased estimated pCR from 20% to 60% in TNBC (5)
- Ongoing Phase III studies in metastatic TNBC:
 - Study of atezolizumab and paclitaxel vs. placebo and paclitaxel in participants with previously untreated locally advanced or metastatic triple negative breast cancer (TNBC) (IMpassion131)
 - Study of pembrolizumab plus chemotherapy vs. placebo plus chemotherapy for previously untreated locally recurrent inoperable or metastatic TNBC (KEYNOTE-355)
- Ongoing Phase III study of atezolizumab in early TNBC:
 - Study to investigate atezolizumab and chemotherapy compared with placebo and chemotherapy in the neoadjuvant setting in participants with early stage triple negative breast cancer (IMpassion031)

1) Adams S. J Clin Oncol. 2016; 34(15 suppl) Abstr 1009
2) Schmid P. Cancer Res. 2017; 77(13 suppl) Abstr 2986
3) Pohlman P. Cancer Res. 2018; 78 Abstr CT028
4) Schmid P. N Engl J Med. 2018 Oct 20
5) Nanda R. J Clin Oncol. 2017; 35(15 suppl) Abstr 506

Eligibility

- Diagnosis of invasive adenocarcinoma of the breast by core needle biopsy
- Primary tumor must be
 - T_2 or T_3 if node negative
 - T_{1C}, T_2, or T_3 if node positive
- Central testing must confirm
 - HER2 negative by ASCO/CAP guidelines
 - ER and PR negative by ASCO/CAP guidelines
- LVEF ≥55%
- Serum TSH and AM cortisol WNL
- Standard exclusions for cardiac disease and for checkpoint inhibitor trials

Stratification Variables

- Size of Primary Tumor
 - 1.1 - 3.0 cm
 - >3.0 cm
- Clinical Nodal Status
 - Negative
 - » Axillary imaging is negative *OR*
 - » Axillary imaging is suspicious or abnormal but FNA/core biopsy negative
 - Positive
 - » FNA or core biopsy of node(s) positive *OR*
 - » Axillary imaging suspicious or abnormal but FNA/core biopsy not done
- AC/EC schedule
 - q2wk
 - q3wk
- Region
 - North America
 - Europe

Schema

TNBC Diagnosed by Core Needle Biopsy with T2-T3 if Node (-) and T1c-T3 if Node (+)

Randomly Assigned

Arm 1	Arm 2
Carbo q3wk x 4 + WP x 12 + Placebo ↓ AC/EC x 4 + Placebo	Carbo q3wk x 4 + WP x 12 + Atezolizumab ↓ AC/EC x 4 + Atezolizumab

SURGERY (lumpectomy or mastectomy) and axillary staging

Arm 1	Arm 2
Placebo to complete 1 Year	Atezolizumab to complete 1 year

NCT03281954

Treatment Doses and Schedule

- Sequence 1 chemotherapy over 12-16 wks as tolerated
 - Carboplatin AUC of 5 q3wks x 4 doses
 - Paclitaxel 80 mg/m^2 IV weekly x 12 doses
- Sequence 2 chemotherapy 4 doses (q2wk vs q3wk per investigator)
 - Doxorubicin (A) 60 mg/m2 IV or Epirubicin (E) 90 mg/m^2 IV
 - Cyclophosphamide (C) 600 mg/m^2 IV
- Atezolizumab 1200 mg or placebo IV every 3 wks initiated with chemotherapy and administered for 1 year with break for surgery

Statistical Considerations

- Sample size: 1,520 pts
- Co-primary endpoints
 - pCR breast and lymph nodes (ypT0/Tis ypN0)
 - Event-free survival (EFS)
- Secondary endpoints
 - pCR breast (ypT0/Tis)
 - pCR breast and lymph nodes (ypT0 ypN0)
 - Positive nodal status conversion rate
 - Overall survival
 - Recurrence-free interval
 - Distant disease-free survival
 - Brain metastases-free survival
 - Toxicity
 - Cardiac safety lead-in study

Cardiac Safety Lead-in Component

- First 60 pts to begin AC/EC
- Troponin T collected prior to AC/EC in Cycle 1 at baseline
- Troponin T collected immediately after AC/EC administration prior to atezo/placebo administration in Cycles 1 and 3
- ECG obtained with each Troponin T collection to allow administration of atezo/placebo without waiting for troponin T result if no change from baseline
- LVEF assessed prior to Cycle 3 of AC/EC
- Cardiac evaluation if troponin T elevated or LVEF declines
- All pts have post op and 18-month LVEF

Rationale for Carboplatin as Component of Chemotherapy Regimen

- GEPARSIXTO TNBC Cohort (1,2)
 - pCR breast and nodes increased from 36.9% to 53.2% (p=0.005) with addition of carboplatin to weekly anthracycline/taxane regimen
 - Carboplatin improved 3-yr DFS (85.8% *v* 76.1%, p=0.035)
- CALGB 40603 (3,4)
 - pCR breast and nodes increased from 41% to 54% (p=0.0029) with addition of q3wk carboplatin AUC 6 to weekly paclitaxel→AC
 - 3-yr EFS was 76% with carboplatin and 71% (p=0.36) without
 - Paclitaxel delivery reduced by requirement to end therapy after 12 wks
- BrighTNess (5)
 - pCR breast and nodes increased from 31% to 57.5% with addition of q3wk carboplatin AUC 6 to weekly paclitaxel→AC
 - Pre-specified analyses by stratification factors demonstrated no differences in pCR by germline BRCA status or schedule of anthracycline administration
 - EFS data not mature
 - Paclitaxel delivery improved by extending therapy to 16 wks if needed

1) von Minckwitz G. Lancet Oncol. 2014; 15:747-756
2) Hahnen E. JAMA Oncol. 2017; 3:1378-1385
3) Sikov, WM. J Clin Oncol. 2015; 33:13-21
4) Sikov, WM. Cancer Res. 2016; 76(4 suppl) Abstract S2-05
5) Loibl S. Lancet Oncol. 2018; 19:497-509

Rationale for Maintaining Placebo Control Post Surgery

- Hypothesis is that cohort receiving atezolizumab will have higher pCR rate, and the increased activity will result in improved EFS
- Imbalance of non-protocol therapy following surgery would compromise integrity of trial for EFS, particularly administration of PD-1/PD-L1 inhibitors
- No evidence additional chemotherapy following Carbo/Taxol→AC will improve outcomes
- CREATE-X was conducted in pts who did not receive neoadjuvant carboplatin

Biospecimen Collections in Support of Correlative Studies

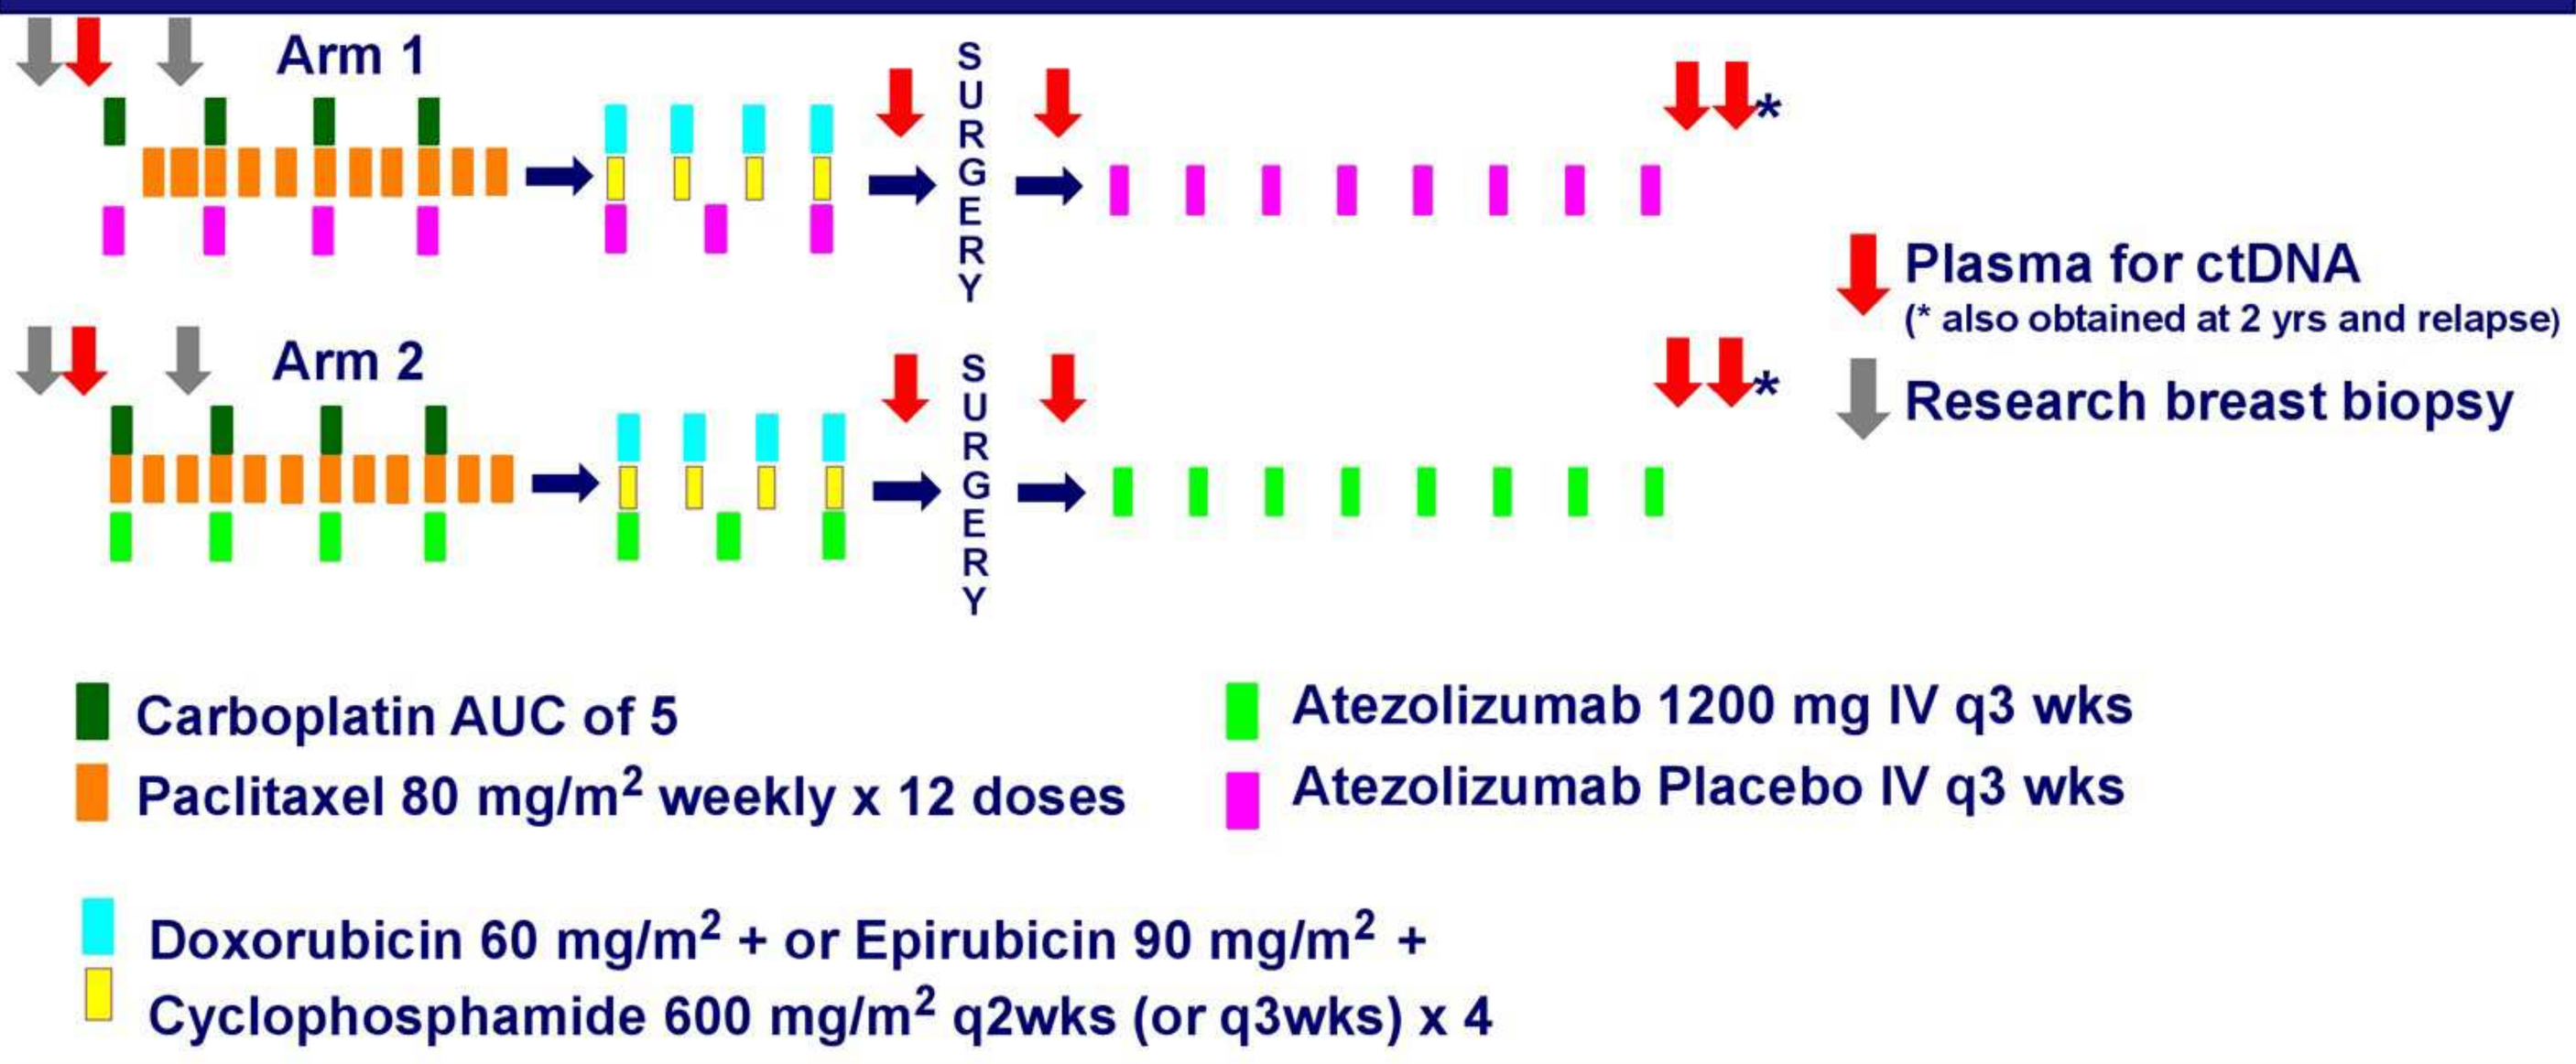

Arm 1
SURGERY
Arm 2
SURGERY
Plasma for ctDNA
(* also obtained at 2 yrs and relapse)
Research breast biopsy
Carboplatin AUC of 5
Paclitaxel 80 mg/m2 weekly x 12 doses
Atezolizumab 1200 mg IV q3 wks
Atezolizumab Placebo IV q3 wks
Doxorubicin 60 mg/m2 + or Epirubicin 90 mg/m2 +
Cyclophosphamide 600 mg/m2 q2wks (or q3wks) x 4

Correlative Studies Aims

- Evaluate expression of PD-L1, and percentages of TILs at baseline and following initial dose of atezolizumab with P/Cb as predictors for pCR and EFS
- Evaluate percentages of TILs in pts with residual breast cancer surgery as predictor for EFS
- Use baseline and on-therapy specimens to explore potential new biomarkers of responses and resistance to neoadjuvant atezolizumab with chemotherapy followed by adjuvant administration of atezolizumab
- Evaluate serial ctDNA as predictive biomarker for pCR and EFS, and as early predictor of recurrence

NSABP B-59/GBG 96-GeparDouze — STATUS

- Academic collaboration between NSABP and GBG with support from Genentech/Roche
- NSABP site activation: December 2017
- GBG site activation: May 2018
- Amendment to add PD-L1 status as a stratification factor and to allow entry of asymptomatic pts with abnormal TSH and cortisol
- Accrual: 232/1520 pts as of 5-21-19

Abstract No. TPS605

2019 ASCO Annual Meeting May 31– June 4. Data is the intellectual property of the author. Contact cegeyer@vcu.edu for permission to reprint and/or distribute.